SNS-595 Demonstrates Clinical Activity and Dose-Proportional Pharmacokinetics (PK) in Two Phase 1 Clinical Studies

H. Hurwitz[1], M. Gordon[2], R. Advani[3], S. Ebbinghaus[4], D. Mendelson[2], H. Wakelee[3], U. Hoch[5], J. Silverman[5], N. Havrilla[5], D. Adelman[5];

[1]Duke University, Durham, NC, [2]Premiere Oncology, Scottsdale, AZ, [3]Stanford University, Stanford, CA, [4]Univ of Arizona, Tucson, AZ, [5]Sunesis Pharmaceuticals, Inc, So. San Francisco, CA

ABSTRACT (Updated May 2006)

Background: SNS-595 is a novel naphthyridine analog with broad and potent activity in preclinical models.

Methods: SNS-595 was administered to patients (pts) with advanced solid cancers as an IV infusion over 10 minutes on 2 schedules: (A) qwk x3/1 week off, and (B) q3wks. Pt eligibility included refractory solid tumors and adequate organ function.

Results: In study A, 21 pts were treated in 6 cohorts (dose range 3-24 mg/m2/wk). In study B, 41 pts were treated in 9 cohorts (dose range 3-75 mg/m2/wk). After the MTD in heavily pretreated (HP) pts was found in study B, dose escalation in minimally pretreated (MP) pts was evaluated. The median ages were 61 yrs (A) and 59 yrs (B), sex 12F/9M (A), 16F/25M (B), all pts had baseline ECOG PS 0-2. Neutropenia was the dose limiting toxicity (DLT) for both studies. The MTD for A was 15 mg/m^2; the MTD for B was 48 mg/m2 for HP and 60 mg/m2 for MP pts. For both studies 2 pts had grade 4 thrombocytopenia; non-hematologic toxicities were mostly grade 1/2. In A, PK were assessed on Days 1 and 15 (after the 1st and 3rd doses); exposure increased linearly over an 8-fold dose range (1.6-15 µghr/mL), CL, Vss and T1/2 averaged 2 L/hr/m2, 49 L/m2, 18 hr, respectively, and did not change from Day 1 to 15. In B, PK were assessed on Day 1 after the 1st dose; exposure increased linearly over the 24-fold dose range (1.1-46 µghr/mL), CL, Vss, and T1/2 averaged 2 L/hr/m2, 53 L/m2, and 21 hrs, respectively. For A, best responses were 1 PR and 6 SD (range 16-24 wks); for B, best responses were 1 PR and 11 SD (range 18-58 wks).

Conclusion: SNS-595 was well tolerated and showed clinical activity with both qwk and q3wk dosing. The DLT was non-cumulative neutropenia. SNS-595 demonstrated remarkably predictable PK, with low inter- and intra-pt variability. Based on these data, phase 2 studies of SNS-595 as monotherapy are ongoing, and are planned in combination therapy.

BACKGROUND

SNS 595 is a novel naphthyridine analog, a class of compounds not previously used for cancer treatment. SNS-595 causes discrete double-strand DNA breaks in cells in S phase resulting in rapid apoptosis and an arrest in G2 phase of the cell cycle.

STUDY OBJECTIVES

- To determine the safety and tolerability of IV SNS-595 given either weekly x3 in a 28-day schedule or every 3 wks
- To assess the PK profile of SNS-595 after single and repeat administration
- To define a recommended dose regimen for subsequent phase 2 efficacy studies
- To obtain preliminary objective tumor response data

METHODS AND SCHEMA

DLT definition

- ANC ≤500 for ≥ 7d or febrile neutropenia
- Platelet nadir <25000 or bleeding
- Non-hematologic AE ≥Grade 3 (CTCAE v3.0)
- AE requiring >14 days dose delay

MTD Definition

- Dose level below dose where ≥ 2 of 6 pts experienced DLT

Heavily pretreated (HP), Minimally pretreated (MP) definition

(Tolcher et al, JCO 2001; 19:2937-2947)

- **HP**: Pts previously received >6 courses of an alkylating agent chemotherapy or >2 courses of platinum, mitomycin-C or any nitrosourea, or XRT to >25% of bone
- **MP**: Those pts not fulfilling the HP definition

Study Schema

- Drug administered by IV bolus, either every 3 wks or weekly x 3 followed by 14 days of observation
- Starting dose: 3 mg/m2; dose escalation in cohorts of 3: dose doubled to first ≥Grade 2, related AE or abnormal lab value, then by a modified Fibonacci schema
- No mitomycin-C, BCNU, nitrosourea drugs; no MAb therapy w/in 42 d

PATIENTS

Table 1: Patient Demographics

	qwk x3	q3wk	total
n (# treated)	21	41	62
Sex			
Male	9 (43%)	25 (61%)	34 (55%)
Female	12 (57%)	16 (39%)	28 (45%)
Ethnic Background			
Asian	2 (10%)	1 (2%)	3 (5%)
Black	2 (10%)	4 (10%)	6 (10%)
Hispanic	0	1 (2%)	1 (2%)
Native Hawaiian/ Pacific Islander	0	2 (5%)	2 (3%)
White	17 (81%)	33 (81%)	50 (81%)
Age (yrs)			
Mean	59.3	58.5	58.8
Median	61	59	60
Range	19-81	33-79	19-81
Previous Therapies			
MP	9 (43%)	17 (41%)	26 (42%)
HP	12 (57%)	24 (59%)	36 (58%)

Table 2: Tumor Types

	qwk x3	q3wk	total
n (# treated)	21	41	62
Ovarian	1	9	10
Colon	3	6	9
NSCLC	0	6	6
Pancreas	3	2	5
Renal	1	4	5
Melanoma	1	3	4
Adeno CA (origin unk)	0	3	3
Breast	2	0	3
Sarcomas	0	3	3
Cholangiocarcinoma	1	1	2
Mesothelioma	2	0	2
Neuroendocrine	1	1	2
Bladder	0	1	1
Leiomyosarcoma	1	1	1
Liposarcoma	1	0	1
Müllerian	0	1	1
Nasopharyngeal	1	0	1
Salivary Gland	1	0	1
Small Cell Lung	1	0	1
Spindle Cell	1	0	1

SAFETY DATA

SNS-595 is well tolerated

- Neutropenia was the primary Grade 3/4 toxicity observed and was the dose limiting toxicity for both studies. Neutropenia was of short durations and was not cumulative.
- Very low incidence of other hematologic toxicities
- Very low incidence of non-hematologic toxicity

Table 3: Frequent (>10%pts) Adverse Events

Body System	preferred term	qwk x3	q3wk	total
	preferred term	n=21	n=41	n=62
Cardiac Disorders				
	edema	0/4*		
Gastrointestinal Disorders				
	abd pain	1/5	0/8	1/13 (8%)
	constipation	0/7	1/12	1/19 (5%)
	diarrhea	0/4	0/8	0/12 (0%)
	nausea	0/8	2/26	2/34 (6%)
	vomiting	0/5	1/17	1/22 (5%)
General Conditions				
	fatigue	1/4	1/13	2/17 (12%)
Metabolism & Nutrition Disorders				
	anorexia		0/7	0/7 (0%)
Musculoskeletal & Connective Tissue				
	backpain		0/7	0/7 (0%)
	pain in extremity	0/5		
Nervous System Disorders				
	dizziness		0/6	0/6 (0%)
	headache	0/3	0/8	0/11 (0%)
Skin & Subcutaneous Tissue Disorders				
	alopecia		0/8	0/8 (0%)

*no. pts with Grade ≥ 3 / no. pts with any Grade

Table 4: Hematologic Effects

	qwk x3	q3wk	total
	n=21	n=41	n=62
Grade 4 Neutropenia (ANC<500/mm^3)	0	10 (24%)	10 (16%)
Febrile Neutropenia	0	1 (2%)	1 (2%)
Grade 4 Thrombocytopenia (<25,000/mm^3)	0	2 (5%)	2 (3%)

Table 5: SAE's Possibly Related to Study Drug

SAE preferred term	CTCAE v3.0 Grade	
1pt for each of the following	qwk x3	q3wk
Sepsis	not observed	Grade 3
Vomiting	not observed	Grade 3
Pneumonia	not observed	Grade 3
Febrile Neutropenia	not observed	Grade 2
Pancytopenia	not observed	Grade 4
Thrombosis	not observed	Grade 2

PHARMACOKINETICS

SNS-595 shows highly reproducible pharmacokinetics

- Low inter-patient variability
- Dose dependent inc. in exposure & no change in CL or Vss with dose & schedule
- No accumulation or change in pharmacokinetic parameters after repeat dosing

Table 6: Average of wk 1 and wk 3 Pharmacokinetic Parameters

	qwk x3, Week 1	qwk x3, Week 3	q3wk
n	20	16	39
Dose Range (mg/m^2)	3 - 24	3 - 18	3 - 75
AUC_{inf} Range (µg•hr/mL)	2 - 15	2 - 7	1 - 46
$T_{1/2}$ (hr) ± SD	22 ± 11	19 ± 8	21 ± 5
Cl_{obs} (L/hr/m^2) ± SD	1.9 ± 0.7	2.2 ± 0.9	2.0 ± 0.4
V_{ss} (L/m^2) ± SD	48 ± 12	47 ± 8	53 ± 4

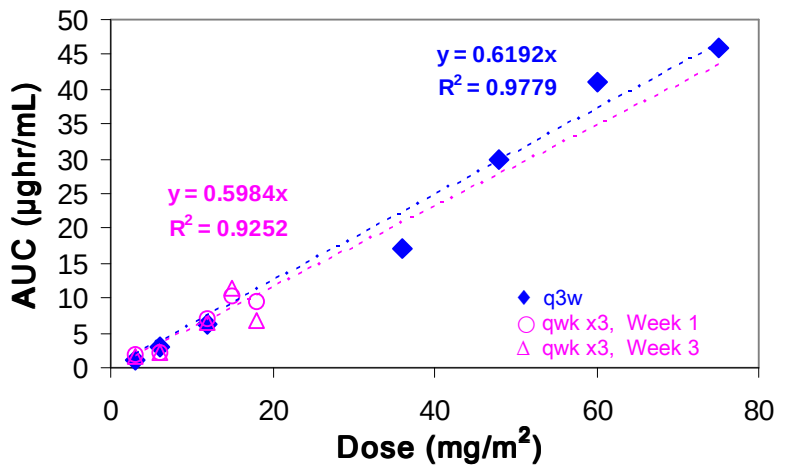
AUC (µghr/mL)
Dose (mg/m2)
y = 0.6192x
R2 = 0.9779
y = 0.5984x
R2 = 0.9252
q3w
qwk x3, Week 1
qwk x3, Week 3

Figure 1: Dose Linearity in Studies A and B. Plot of AUC against doses administered qwk x3 (pink circles week:1, pink triangles: week 3) and q3w (blue diamonds)

EVIDENCE OF ACTIVITY

SNS-595 shows evidence of clinical activity in endstage refractory patients

- 2 patients achieved Partial Responses (PR), 1 each: ovarian, mesothelioma
- 17 of the remaining 59 patients had Stable Disease (SD) while remaining on therapy for at least 16 wks: 2 (NSCLC and neuroendocrine) with SD exceeding 1 year
- Of the 10 ovarian cancer patients enrolled, 4 had at least SD and continued on SNS-595 for 18 to 33 wks

Table 7: Details on Prolonged Stable Disease

(mg/m^2)	Scheduled	Tumor Type	Weeks on	Best
6	qwk x3	Renal Cell	16	SD
12	qwk x3	Leiomyosarcoma	16	SD
		Melanoma	16	SD
15	qwk x3	Mesothelioma	28	PR
		Mesothelioma	18	SD
		Nasopharyngeal	16*	SD
24	qwk x3	Salivary Gland	24	SD
3	q3wk	Lung	18	SD
6	q3wk	Renal Cell	18	SD
12	q3wk	Lung	53*	SD
24	q3wk	Adenocarcinoma (unknown origin)	18	SD
36	q3wk	Ovarian	18	SD
		Colon	33	SD
48	q3wk	Ovarian	24	PR
		Ovarian	30	SD
		Lung	46	SD
60	q3wk	Ovarian	33	SD
		Neuroendocrine	58*	SD
75	q3wk	Müllerian	46*	SD

* Patient still on study

Table 8: Details of Partial/Minor Responses (PR/MR)

Best Response	Tumor	Criterion	Baseline	C2	C4	C6
PR	Ovarian	CA125 (U/mL)	467	272	176	120
		P-aortic node (cm)	1.7	1.6	1.5	1.3
		Aortic node (cm)	2.4	2.0	2.2	2.0
		Iliac node (cm)	5.5	5.0	4.7	3.9
PR	Mesothelioma	Posterior hemithorax (cm)	2.5	2.5	1.6	1.6
		Pleural nodule in major fissure (cm)	1.2	1.2	1.0	-
SD (MR)	Nasopharyngeal	Rt upper lobe nodule 1 (cm)	2.3	1.5	-	-
		Rt upper lobe nodule 2 (cm)	1.5	1.3	-	-
		Left upper lobe nodule (cm)	1.1	1.0	-	-
		Lingular nodule	1.6	1.5	-	-
SD (MR)	Ovarian	CA125 (U/mL)	567	811	419	274
		Liver met (cm)	7.4	6.9	5.7	-
		Aortic node (cm)	1.7	1.5	1.2	-
		P-aortic node (cm)	1.8	1.1	1.1	-
SD (MR)	Ovarian	CA125 (U/mL)	50	18	16	15
		Rectosigmoid (cm)	2.5	0.0	0.0	0.0
		3 mets unchanged				
SD (MR)	Müllerian	RML Lung (cm)	3.9	3.9	3.7	2.4

CONCLUSIONS AND FUTURE DIRECTIONS

In two phase 1, dose-escalating studies, SNS-595 demonstrates good safety profiles and evidence of anti-tumor activity with either of two administration schedules. Its pharmacological characteristics suggest dosing schedule flexibility as a single agent or in combination therapy.

✓**SNS-595 is well tolerated when given as an IV bolus weekly x 3 and once every 3 weeks**
- Neutropenia was the most common AE, but was of short duration and non-cumulative. Other toxicities were uncommon and usually mild.
- The recommended phase 2 dose for solid tumors is 48 mg/m^2 for q3wk dosing and 15 mg/m^2 for qwk x3 dosing

✓**SNS-595 shows evidence of clinical activity: 2 PR and 17 patients with stable disease ≥ 16 wks**

✓**SNS-595 shows highly reproducible pharmacokinetics**
- Low inter-individual variability and no changes with repeated dosing

✓**Phase 2 trials in NSCLC, SCLC, and a phase 1 study in acute leukemias in progress; additional phase 2 indications planned**
- Favorable PK and safety profile combined with low potential for drug-drug interactions ideal for pursuing combination studies