PHARMACOKINETIC/PHARMACODYNAMIC CORRELATION WITH CLINICAL RESPONSES IN A PHASE 1 STUDY OF PATIENTS WITH RELAPSED/REFRACTORY ACUTE LEUKEMIAS TREATED WITH SNS-595

Michelle R. Arkin[1], Rebecca Ricklis[2], Jeffrey Lancet[3], Frank Giles[4], Ute Hoch[1], Tarra Fuchs-Knotts[1], David Stockett[1], Nguyen Tan[1], Matt Suster[1], Don Young[1], Glenn Michelson[1], Judith A. Fox[1], Judith E. Karp[2]

[1]Sunesis Pharmaceuticals, Inc.; [2]Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins; [3]H. Lee Moffitt Cancer Center and Research Institute; [4]MD Anderson Cancer Center

Introduction and Background

- **SNS-595 is a novel naphthyridine derivative that causes highly selective, replication-dependent DNA-damage, irreversible G2 arrest, and rapid onset of apoptosis**
- **The complex mechanism of action of SNS-595 includes non-classical inhibition of topoisomerase II that is clearly differentiable from classic inhibitors such as etoposide and doxorubicin**
- **SNS-595 is in Phase 1 and 2 clinical trials in hematologic and solid malignancies**
- **Preliminary clinical, pharmacokinetic (PK) and correlative results from an on-going escalating-dose phase 1 trial of SNS-595 in refractory acute leukemias are presented**

Mechanism of Action of SNS-595

SNS-595

↓

Double-strand break induced during replication

Cell arrest and death

Damage repaired by NHEJ

Aims

- Determine the safety and tolerability of escalating doses of SNS-595 Injection in patients with advanced leukemias
 - **Schedule A:** Qwk IV administration of SNS-595 Injection (Days 1, 8, and 15) for 3 doses/cycle
 - **Schedule B:** BIW IV administration of SNS-595 Injection (Days 1, 4, 8, and 11) for 4 doses/cycle
- Characterize SNS-595 pharmacokinetics (PK) in this patient population
- Evaluate pharmacodynamic (PD) biomarkers of mechanism-based activity
- Explore potential relationships between between PK, PD and clinical outcome.
- Obtain preliminary assessments of anti-leukemia activity
- Define a recommended dose regimen for future phase 2 studies

Major Entry Criteria and Laboratory Correlates

- Relapsed or refractory leukemia
 - $\leq$ 3 induction/re-induction regimens for either AML or ALL
 - Adequate renal and hepatic function
- PK samples were collected at timepoints up to 96 hr post dose on Day 1 (schedule A) and Days 1, 4, 8, 11 (schedule B)
- Pre- and post-dose blood samples were obtained on Day 0, Day 1 (2 – 4 hours post-dose), Day 8 (predose) and Day 8 (2 – 4 hours post-dose) from a subset of patients to examine for evidence of:
 - DNA damage by H2AX phosphorylation (γH2AX)
 - Apoptosis by γH2AX and PARP cleavage
 - Cell-cycle response

SAFETY DATA

Table 1: Grade 3 or 4 Adverse Events with > 5% Incidence

	Schedule A (qw x3)					Schedule B (biw x4)						
assigned dosage (mg/m^2)	18	27	38	50	60	9	14	19	25	30	40	total
# Patients Treated	3	7	3	8	4	4	4	3	3	2	NA	41
# Pts Reporting grade 3 or 4 AEs	**2**	**5**	**1**	**5**	**1**	**4**	**4**	**2**	**3**	**2**		**29**
Febrile neutropenia	2	3	1	4	1	1	0	1	2	0		15
Thrombocytopenia	1	4	0	1	0	1	2	0	0	2		11
Neutropenia	2	1	0	2	0	0	2	1	1	1		10
Anaemia	0	1	0	0	0	0	1	0	0	1		3
Leukocytosis	0	2	0	0	0	0	1	0	0	0		3
Pancytopenia	0	0	0	1	0	0	0	1	1	0		3
Pyrexia	1	0	0	0	0	2	0	1	0	0		4
Pneumonia	1	1	0	1	1	1	1	1	1	0		8
Bacteraemia	1	0	0	0	1	0	0	0	1	0		3
Catheter related infection	1	0	0	0	0	0	1	0	0	0		2
Infection	0	1	0	0	0	1	0	0	0	0		2
Sepsis	0	0	0	2	0	0	0	0	0	0		2

DLT Definition:

- NCI CTCAE Grade 4 hematologic event(s) of neutropenia or thrombocytopenia occurring through Cycle 1 Day 29 that are assessed as clinically significant and related to study drug and that persist in the absence of viable leukemia beyond 8 weeks after the Cycle 1 Day 1 dose
- $\geq$ NCI CTCAE Grade 3 nonhematologic event(s) occurring through Cycle 1 Day 29 that are assessed as clinically significant and related to study drug, regardless of duration.

To date, there has been one hematologic DLT (prolonged neutropenia, at 50 mg/m^2 qw) and no nonhematologic DLT.

Clinical Responses

Table 2: Bone Marrow Responses in AML

Dose (mg/m^2)	Age/ sex	Relapse/ refractory	# prior regimens	Cytoge-netic risk	Blast % at diagnosis	Lowest Blast %	Clinical Response
50 qw x 3							
2016	66/M	Rel	3	NA[a]	12	0	NR[b]
2017	77/M	Rel+Ref	4	poor	56	0	NR
2019	75/F	Rel	1	Intermed.	20	0	NR
2021	66/M	Rel	3	Intermed.	65	3	**CRp**
60 qw x 3							
2024	74/M	Ref	3	poor	63	3	CRi[c]
2025	59/M	Rel+Ref	2	Intermed.	80	15	PR[d]
40 biw x 4							
2118	76/F	Rel	2	Intermed.	20	0	**CR**

[a]NA = not available; [b]NR = no response; [c]blood showed trilineage hematopoiesis and normal cytogenetics; patient died from infection before complete count recovery; [d]PR = partial response; > 50% reduction in blasts to < 25%, > 5%;. **Pts in RED are still on study.**

PK Parameter is Associated With Clinical Activity

Clinical Responses Appear To Correlate With Weekly Time Above 1 μM Threshold Concentration

SNS-595 Plasma Concentration-Time Profiles

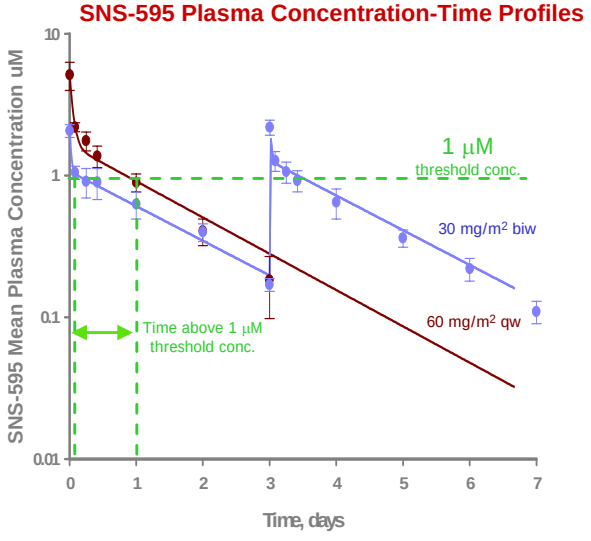
SNS-595 Mean Plasma Concentration uM
10
1
0.1
0.01
0
1
2
3
4
5
6
7
Time, days
1 µM
threshold conc.
30 mg/m² biw
60 mg/m² qw
Time above 1 µM
threshold conc.

- CRs observed when 1 μM plasma concentrations were sustained for >20 hr (Table 2)
- Schedule A, average time above 1 μM increased to >20 hr at doses of >50 mg/m^2
- Schedule B, >20 hr above 1 μM was observed in only 1 patient at 40 mg/m^2 ; patient had a CR
- Total weekly AUC and dose did not correlate with response

Table 3: PK Summary

Single dose (mg/m^2)	Time above 1 μM per week (hr, mean ± SD)	AUC per week (μM*hr, mean ± SD)
Schedule A		
18	0.8 ± 1	20 ± 4
27	10 ± 8	44 ± 13
38	10 ± 4	37 ± 12
50	22 ± 8	68 ± 19
60	21 ± 5	69 ± 12
Schedule B		
9	<1hr	28 ± 16
14	1 ± 1	30 ± 7
19	2 ± 2	49 ± 11
25	6 ± 3	67 ± 13
30	16 ± 9	90 ± 11
40	14 ± 12	80 ± 21

- $T_{1/2}$ = 24 ± 7 hrs
- Cl_{obs} = 2.09 ± 0.65 L/hr/m^2
- V_{ss} = 64 ± 18 L/m^2
 are constant throughout the dose range
- Low interpatient variability and dose-linear exposure
- No accumulation with repeat dosing, and no systematic change in PK parameters

Evidence For Mechanism-Based Activity in Peripheral Blasts

DNA Damage And Apoptosis Are Observed At 60 mg/m^2 Qwk Pt 2024 Shows Greater Apoptotic And Bone Marrow Responses than Pt 2025

DNA damage → γH2AX → Apoptosis → γH2AX + cleaved PARP

FACS analysis of γH2AX

	% cells with γH2AX	
time	**2024**	**2025**
day 0	13	40
day 1, 2-4 hrs post	40	27
day 8, pre	49	63
day 8, 2-4 hrs post	71	66

Western analysis of PARP

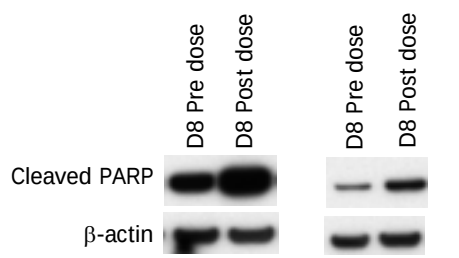
2024
2025
D8 Pre dose
D8 Post dose
D8 Pre dose
D8 Post dose
Cleaved PARP
β-actin

CORRELATION BETWEEN PK and PD

Hours above threshold concentration of 1 μM per week

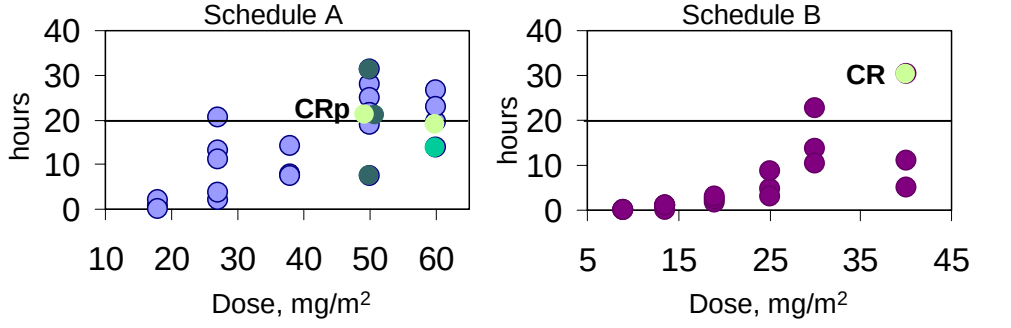
Schedule A
Schedule B
hours
40
30
20
10
0
CRp
CR
10
20
30
40
50
60
5
15
25
35
45
Dose, mg/m²

Patients with > 95% reduction in BM blasts

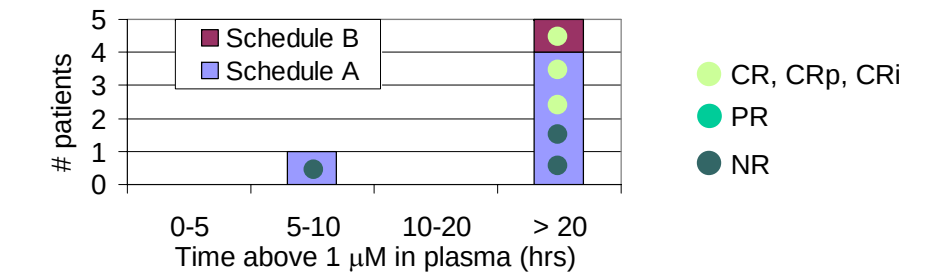
patients
5
4
3
2
1
0
Schedule B
Schedule A
0-5
5-10
10-20
> 20
Time above 1 µM in plasma (hrs)
CR, CRp, CRi
PR
NR

Clinical Responses Are Observed When Plasma Concentrations of SNS-595 Exceed 1 μM for >20 hrs/week

- 1 μM ~ IC_{90} for antiproliferative activity of SNS-595 in two AML cell lines
- "Time above" association is consistent with cell culture data showing that >5 hr exposure is needed to convert DNA damage into apoptosis

CONCLUSIONS AND FUTURE DIRECTIONS

SNS-595 demonstrates:

- Clinically important activity, including CR and CRp, when administered on either a weekly or biweekly schedule
- Potential for flexibility when dosed in combination with other anti-leukemia agents
- Responses at tolerable doses
 - Allowing multiple cycles of SNS-595 without reduction in dose
 - No dose-limiting gastrointestinal toxicities, including mucositis, yet observed at clinically active doses
- DNA damage responses, consistent with its mechanism of action
- A relationship between sustained threshold levels and clinical responses

- Dosing can be rationally selected to take into advanced clinical studies
- Dose escalation continuing (72 mg/m^2 schedule A; 50 mg/m^2 schedule B); DLT not yet reached
- Next Steps:
 Initiate a Phase 1b study of SNS-595 in combination with cytarabine in patients with advanced acute leukemias later this year